#382

A DOUBLE-BLIND, PLACEBO-CONTROLLED CLINICAL PHARMACOLOGICAL STUDY OF TAS-303 IN FEMALE PATIENTS WITH STRESS URINARY INCONTINENCE

Yono M[1], Kimura M[2], Inoue Y[2], Furukawa K[2], Ito K[3], Tsuruya K[4], Nagaoka M[4], Hoashi K[4], Irie S[2]

1. Department of Urology, Nishi-Kumamoto Hospital, SOUSEIKAI, Kumamoto, Japan,

2. Department of Clinical Pharmacology, Nishi-Kumamoto Hospital, SOUSEIKAI, Kumamoto, Japan,

3. Department of Public Health, Osaka City University Graduate School of Medicine, Osaka, Japan,

4. Clinical Development Division II, Taiho Pharmaceutical Co., Ltd., Tokyo, Japan

Hypothesis / aims of study

TAS-303, a selective noradrenaline reuptake inhibitor, is currently at the stage of Phase II development for stress urinary incontinence (SUI) in Japan.

In animal studies, TAS-303 has been shown to increase urethral basal pressure in normal rats, and to increase the reduced leak point pressure (LPP) in a rat vaginal distension (VD) model.

The aim of this study was to evaluate pharmacological effect, safety and pharmacokinetics of TAS-303 in female patients with SUI.

Objective

Primary objective

- To explore the effects of TAS-303 on urethral pressure in patients with SUI when given as a single oral dose of TAS-303

Secondary objective

- To evaluate the safety and the pharmacokinetics of TAS-303 given as a single oral dose in SUI patients

Material and Methods

A double-blind, single-dose, placebo-controlled 2-period crossover study.

Patients had to have the predominant symptom of SUI with ≥2 incontinent episodes per week and with moderate to severe leakage on 1-hour pad test. The PK Parameters were analyzed using Phoenix® WinNonlin® and other data were analyzed using SAS Ver. 9.2.

This study was conducted in accordance with Good Clinical Practice (GCP) Guidelines and Declaration of Helsinki. The protocol and informed consent were approved by the Institutional Review Boards of Hakata Clinic in Japan. Informed consent was obtained from all the participants included in the study. (Registration number: NCT02562807)

Results

Figure 1. Disposition of subjects

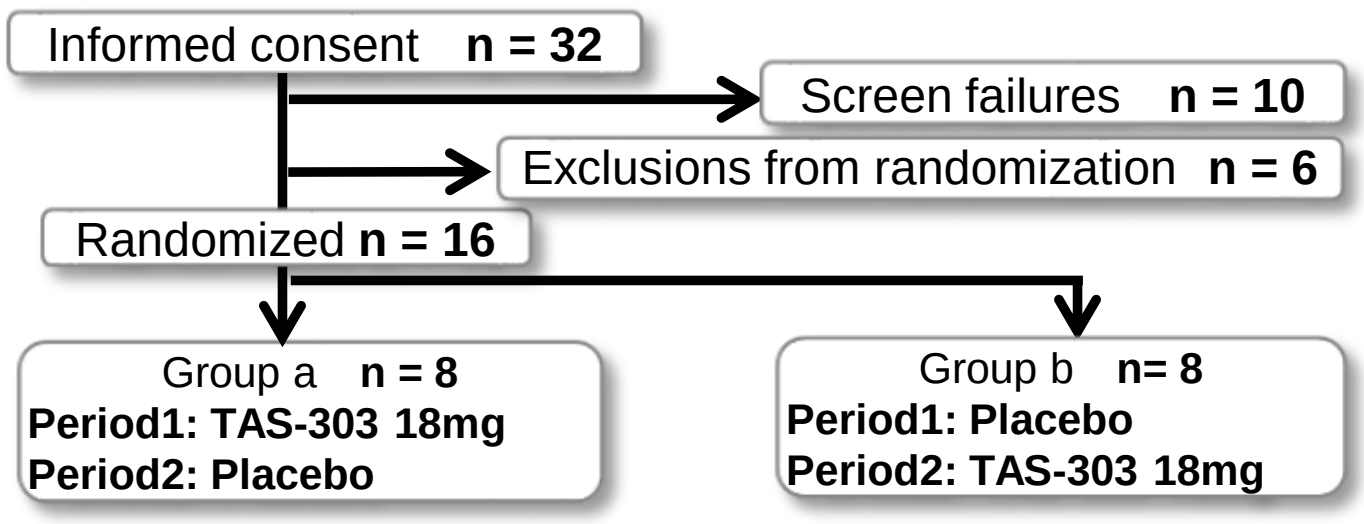
Informed consent n = 32
Screen failures n = 10
Exclusions from randomization n = 6
Randomized n = 16
Group a n = 8
Period1: TAS-303 18mg
Period2: Placebo
Group b n= 8
Period1: Placebo
Period2: TAS-303 18mg

Table 1. Patient Characteristics

Variable		N
Gender ,Ethnicity	Female, Japanese	16 (100.0%)
Type of Urinary Incontinence	Stress Urinary Incontinence	10 (62.5%)
	Mixed Urinary Incontinence	6 (37.5%)
Age (years)	Median [Min, Max]	53.0 [23, 62]
BMI (kg/m²)	Median [Min, Max]	20.69 [18.5, 29.7]

Figure 2. Mean changes in MUCP at 6 hr Post-dose

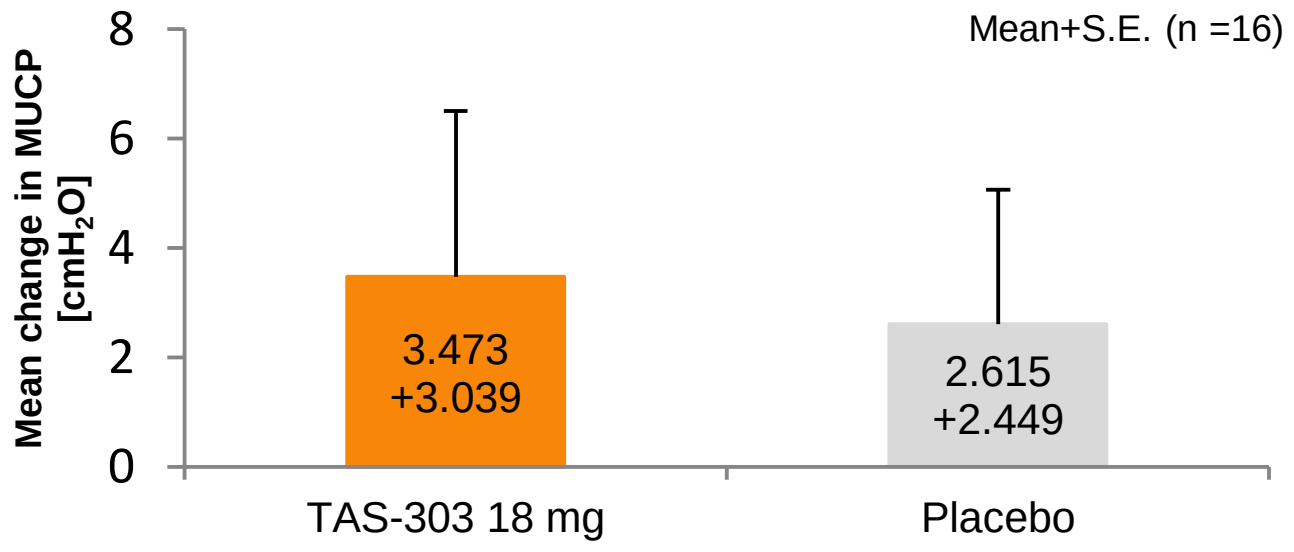
Mean+S.E. (n =16)
Mean change in MUCP [cmH2O]
8
6
4
2
0
3.473
+3.039
2.615
+2.449
TAS-303 18 mg
Placebo

The mean change+S.E. in MUCP at 6 hr post-dose was 3.473 + 3.039 for TAS-303 and 2.615 + 2.449 for placebo, was slightly higher for TAS-303, but with no significant difference between TAS-303 and placebo.

Figure 3. By-Subject Display MUCP Value at 6 hr Post-dose

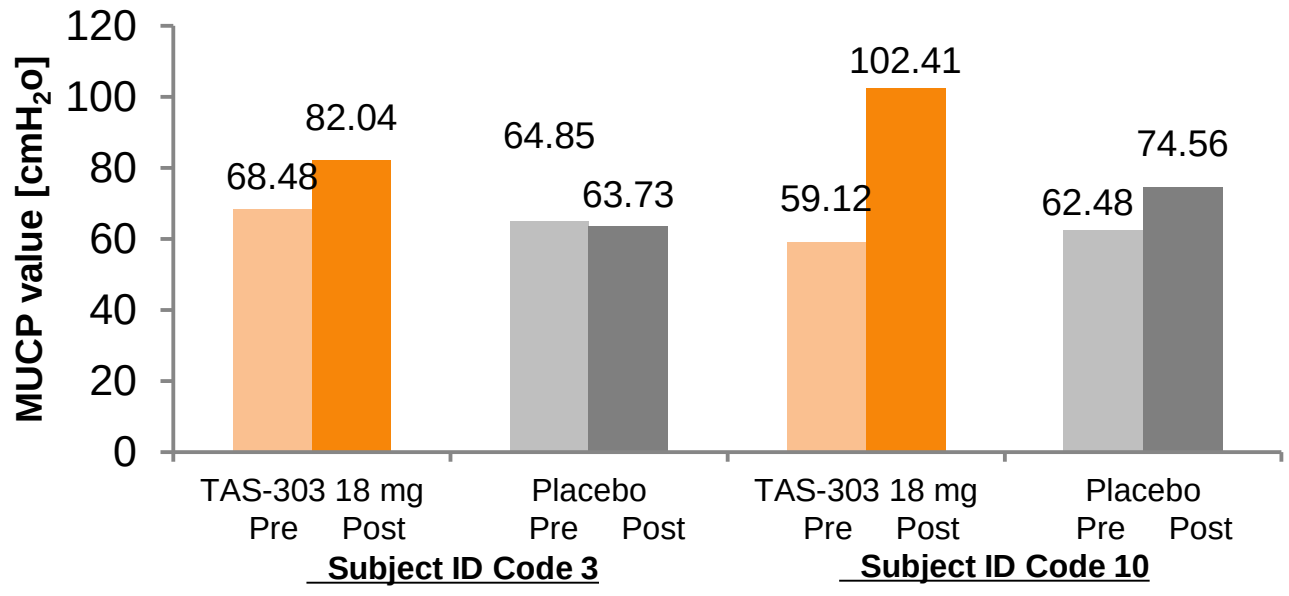
MUCP value [cmH2o]
120
100
80
60
40
20
0
68.48
82.04
64.85
63.73
59.12
102.41
62.48
74.56
TAS-303 18 mg
Pre Post
Placebo
Pre Post
TAS-303 18 mg
Pre Post
Placebo
Pre Post
Subject ID Code 3
Subject ID Code 10

In 2 of the patients, the MUCP increased by more than 10 cmH_2O after administration of TAS-303. Furthermore, one of these 2 patients had particularly high MUCP of more than 100 cmH_2O with TAS-303. Meanwhile, no such changes in MUCP were found in any patients after administration of placebo.

Figure 4. Mean change in MUCP at 6 hr post-dose in each urethral region (proximal, middle and distal)

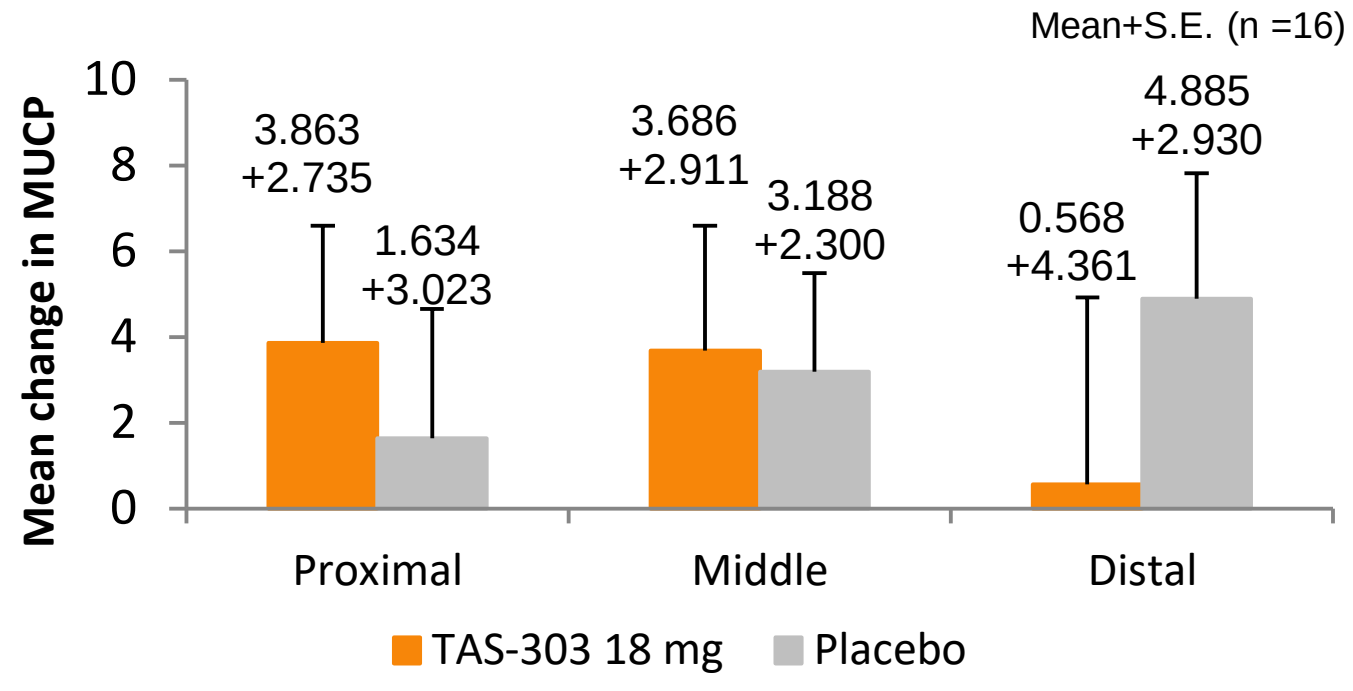
Mean+S.E. (n =16)
Mean change in MUCP
10
8
6
4
2
0
3.863
+2.735
1.634
+3.023
3.686
+2.911
3.188
+2.300
0.568
+4.361
4.885
+2.930
Proximal
Middle
Distal
TAS-303 18 mg
Placebo

There was no significant difference in the change in MUCP between TAS-303 and placebo administration in any sections.

Pharmacokinetics Results

Figure 5. Mean Plasma Concentration-time Profile of TAS-303 in SUI Patient

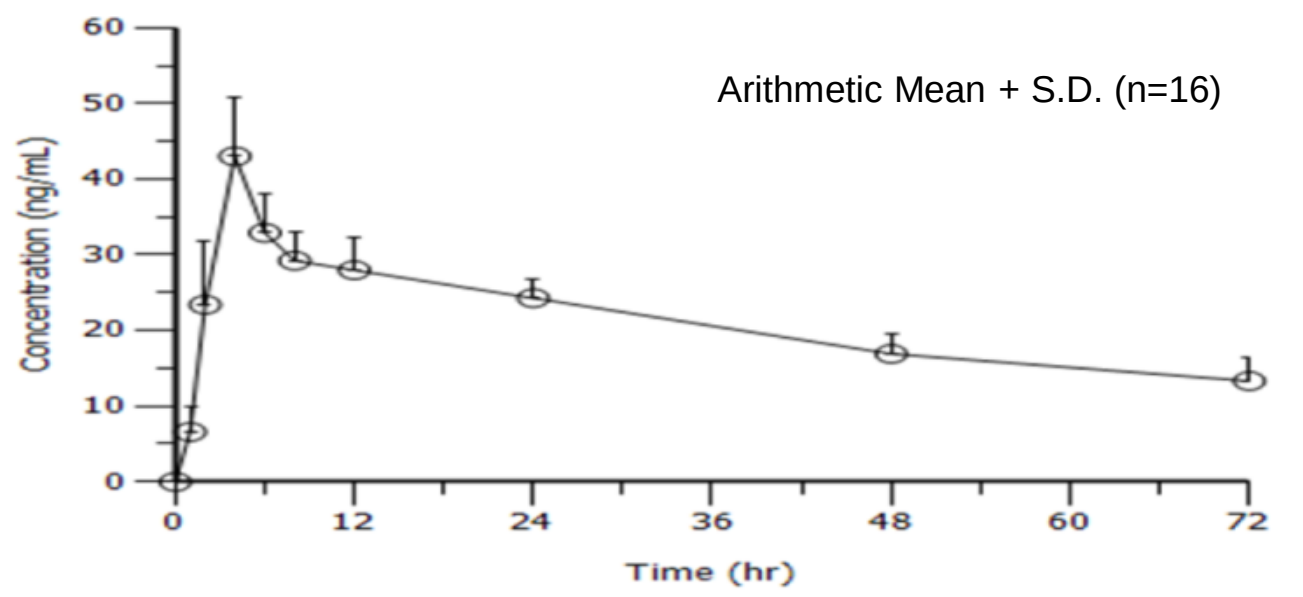
Arithmetic Mean + S.D. (n=16)
Concentration (ng/mL)
60
50
40
30
20
10
0
0
12
24
36
48
60
72
Time (hr)

The geometric mean C_{max}, $AUC_{0\text{-}inf}$ and $t_{1/2}$ of TAS-303 were 42.341 ng/mL, 2677 ng•hr/mL, and 57.76 hr, respectively, and the median t_{max} was 4.0 hr. There appeared to be no clinically significant difference in pharmacokinetic parameters between female SUI patients and healthy adult male subjects.

Safety

Adverse event / Laboratory abnormality

An adverse event of ligament sprain (PT) was reported in 1 of 16 subjects for TAS-303. Ligament sprain developed 5 days after administration of TAS-303, was moderate in severity. The event was assessed as not reasonably related to TAS-303 by the Investigator. No clinically significant abnormal change was observed in laboratory data (hematology, blood chemistry and urinalysis).

Figure 6. Vital sign – Summary Statistics of Blood Pressure

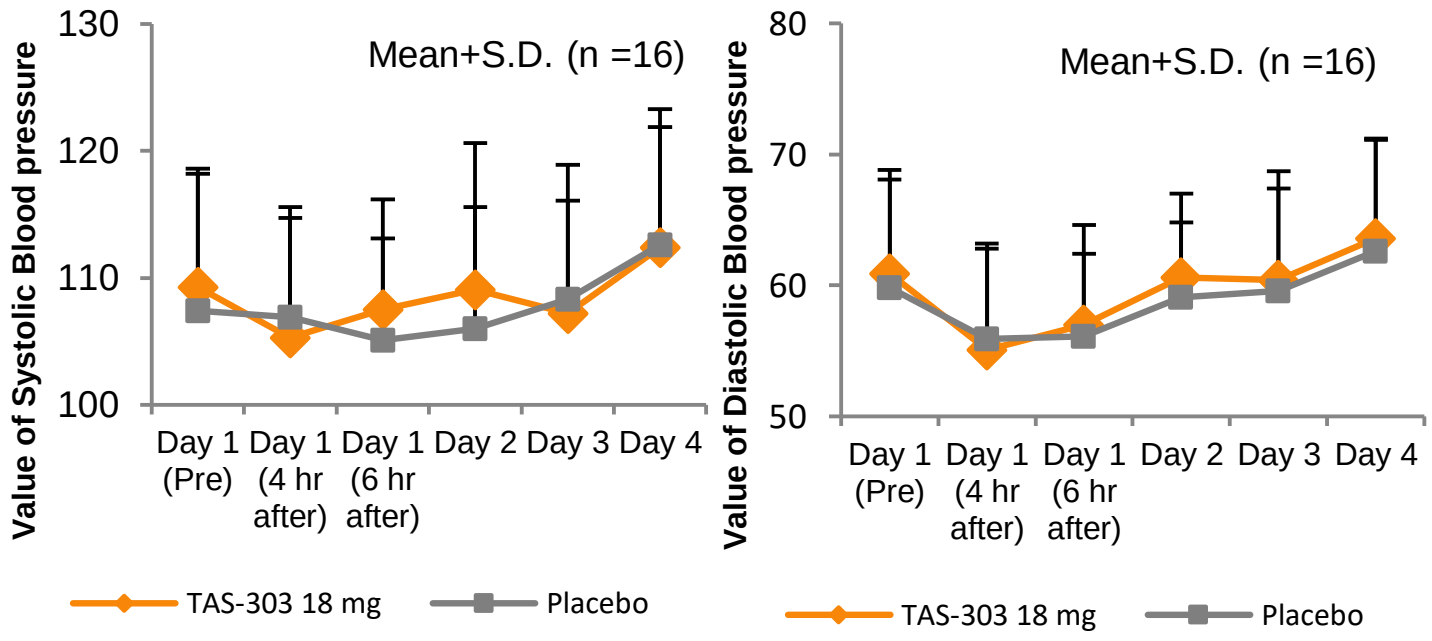
Value of Systolic Blood pressure
Mean+S.D. (n =16)
130
120
110
100
Day 1 (Pre)
Day 1 (4 hr after)
Day 1 (6 hr after)
Day 2
Day 3
Day 4
TAS-303 18 mg
Placebo
Value of Diastolic Blood pressure
Mean+S.D. (n =16)
80
70
60
50
Day 1 (Pre)
Day 1 (4 hr after)
Day 1 (6 hr after)
Day 2
Day 3
Day 4
TAS-303 18 mg
Placebo

No clinically significant abnormal change was observed in blood pressure. No adverse event related to blood pressure was reported. No abnormality in 12-lead electrocardiogram, blood pressure, pulse rate, or body temperature was identified.

Conclusions

Two patients had increased MUCP including 1 patient with particular high MUCP, however, significant difference in MUCP between TAS-303 and placebo in SUI patients was not identified in this study. Considering the limitation of a single-dose study, the efficacy of TAS-303 should not be conclusively determined only by the results of this study. Instead, the efficacy of this drug should be evaluated more accurately in a Phase II study in which the primary endpoint is improvement of SUI symptoms.

No safety concerns were associated with TAS-303 administered to SUI patients as a single dose of 18 mg, indicating the tolerability.

Acknowledgments

This study was funded by TAIHO Pharmaceutical Co., Ltd.

Conflict of interest

Hoashi K. , Nagaoka M and Tsuruya K. are employees of TAIHO Pharmaceutical Co., Ltd. The authors have indicated that they have no other conflict of interest regarding the content of this article.