The effect of mirabegron on bladder blood flow in a rat model of bladder outlet obstruction

Tsuyoshi Majima Yasuhito Funahashi, Shun Takai, Yoshihisa Matsukawa, Tokunori Yamamoto, Momokazu Gotoh

Nagoya University Graduate School of Medicine

ABSTRACT

Introduction

We have evaluated the effects of mirabegron, a β3-adrenoceptor agonist on bladder blood flow in a rat model of bladder outlet obstruction (BOO).

Methods

Adult female Sprague-Dawley rats were divided into 3 groups: sham, BOO, and BOO + mirabegron. In the BOO and BOO + mirabegron groups, rats underwent an operation to establish partial BOO. After the surgery, the BOO + mirabegron group was treated with mirabegron (0.3 mg/kg/h, subcutaneously 14 days. The following experiments were performed after the 14 days-treatment: (1) continuous cystometry; (2) bladder blood flow; (3) hematoxylin-eosin staining of bladder tissue; and (4) malondialdehyde (MDA) measurement in bladder tissue.

Results

(1) continuous cystometry: The BOO group showed significantly higher baseline and peak pressure, more residual urine volume, lower voiding efficiency, and more frequent non-voiding contractions in comparison to the sham group (p = 0.04, < 0.001, 0.007, 0.02, 0.01, respectively).
The BOO + mirabegron group had significantly fewer non-voiding contractions than the BOO group (p = 0.04).
(2) bladder blood flow: The bladder blood flow was significantly different at 0.4 mL intravesical volume among three groups in the following order: sham > BOO + mirabegron > BOO groups
(3) hematoxylin-eosin staining: The bladder tissue in the BOO group had a tendency to contain more hypertrophic detrusor muscle and inflammatory cells when compared to those of the control group, while mirabegron treatment suppressed these histological changes.
(4) MDA measurement: In the BOO group significantly increased MDA levels were detected, as compared to the sham group (p = 0.02). Moreover, significantly decreased MDA was observed in the BOO + mirabegron than the BOO group. (p = 0.04)

Conclusions

Mirabegron treatment significantly improved BOO-induced bladder dysfunction through the amelioration of bladder blood flow.

METHODS

Adult female Sprague-Dawley rats were divided into 3 groups: sham, BOO, and BOO + mirabegron. In the sham group, rats underwent a sham operation, whereas in the BOO and the BOO + mirabegron group, rats underwent an operation to establish partial BOO. After the surgery, the BOO + mirabegron group was treated with mirabegron (0.3 mg/kg/h, subcutaneously) using an osmotic pump for 14 days, whereas the sham and the BOO groups were similarly treated with the vehicle. The following experiments were performed after the 14 days-treatment: (1) continuous cystometry in awake state; (2) measurement of bladder blood flow with a 2D laser blood flow imager (OMEGAZONE); (3) hematoxylin-eosin staining of bladder tissue; and (4) malondialdehyde (MDA) measurement in bladder tissue.

RESULTS

The bladder weight was significantly increased in the BOO group than the sham group (p < 0.001), whereas the BOO + mirabegron group showed significantly decreased bladder weight than the the BOO group (p = 0.02). There was no significant differences in blood pressure among 3 groups.

(1) *Continuous cystometry in awake state*

The BOO group showed significantly higher baseline and peak pressure, more residual urine volume, lower voiding efficiency, and more frequent non-voiding contractions in comparison to the sham group (p = 0.04, < 0.001, 0.007, 0.02, 0.01, respectively). In addition, the BOO + mirabegron group showed significantly higher peak pressure, more residual urine volume and lower voiding efficiency than the sham group. The BOO + mirabegron group had significantly fewer non-voiding contractions than the BOO group (p = 0.04).

(2) *Measurement of bladder blood flow*

There was no significant difference among the three groups, regarding the bladder blood flow with intravesical volume at 0 mL. In contrast, at 0.4 mL intravesical volume, the BOO and BOO + mirabegron group had significantly decreased bladder blood flow than the sham group (p < 0.001 and 0.006), whereas the BOO + mirabegron group showed significantly (p = 0.01) increased bladder blood flow, as compared to the BOO group.

(3) Hematoxylin-eosin staining

The bladder tissue in the BOO group had a tendency to contain more hypertrophic detrusor muscle and inflammatory cells when compared to those of the control group, while mirabegron treatment suppressed these histological changes.

(4) *MDA measurement*

In the BOO group significantly increased MDA levels were detected, as compared to the sham group (p = 0.02). Moreover, significantly decreased MDA was observed in the BOO + mirabegron than the BOO group. (p = 0.04)

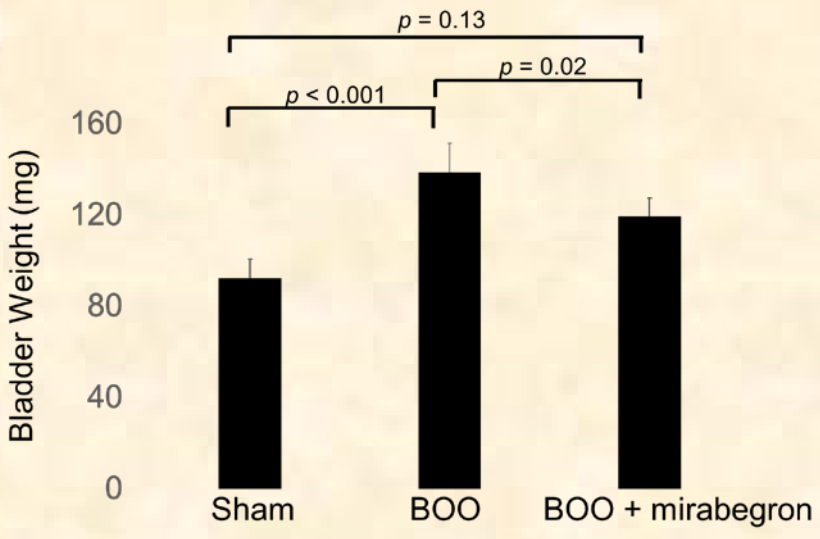
p = 0.13
p = 0.02
p < 0.001
Bladder Weight (mg)
160
120
80
40
0
Sham
BOO
BOO + mirabegron

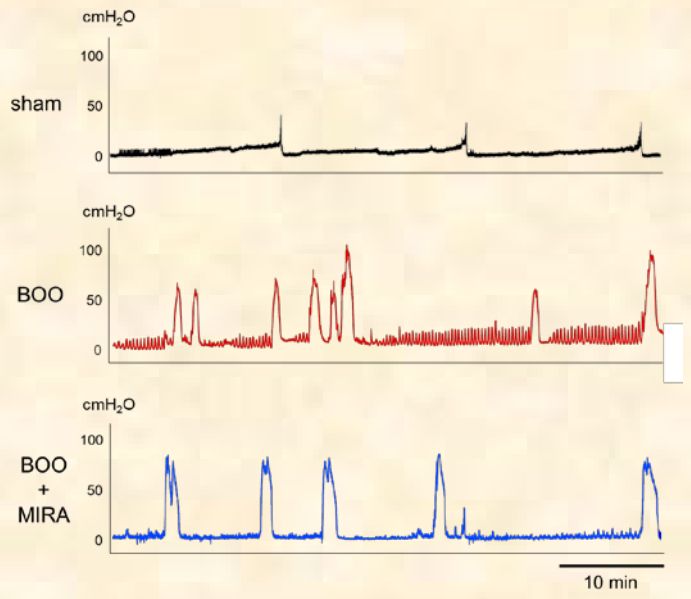
cmH2O
100
50
0
sham
BOO
BOO + MIRA
10 min

		B.P. (cmH_2O)	M.T. (cmH_2O)	P.P. (cmH_2O)	V.V. (mL)	R.V. (mL)	V.E.	Compliance (mL/H_2O)	B.C. (mL)	No of NVCs (n)	Amplitude of NVCs (cmH_2O)
Sham (S)		3.7 ± 0.3	8.9 ± 1.1	36.8 ± 3.6	0.78 ± 0.15	0.02 ± 0.01	0.97 ± 0.01	0.17 ± 0.02	0.80 ± 0.16	1.2 ± 0.8	1.9 ± 1.2
BOO (B)		5.6 ± 0.5	13.7 ± 2.2	81.5 ± 8.4	0.50 ± 0.14	0.28 ± 0.09	0.63 ± 0.10	0.09 ± 0.007	0.77 ± 0.20	6.1 ± 1.5	5.7 ± 1.0
BOO + MIRA (BM)		5.5 ± 0.5	12.6 ± 1.0	70.0 ± 8.0	0.43 ± 0.06	0.31 ± 0.11	0.59 ± 0.10	0.11 ± 0.04	0.71 ± 0.12	2.1 ± 0.7	4.4 ± 1.2
p-value	S vs B	0.04	n.s.	< 0.001	n.s.	0.007	0.02	n.s.	n.s.	0.01	n.s.
	S vs BM	0.05		0.01		0.006	0.001			0.85	
	B vs BM	0.99		0.50		0.97	0.44			0.04	

B.P.: basal pressure, M.T.: micturition threshold, P.P.: peak pressure, V.V.: voided volume, R.V.: residual volume, V.E.: voiding efficiency, B.C.: bladder capacity, NVC: non-voiding contraction, n.s.: not significant, MIRA: mirabegron

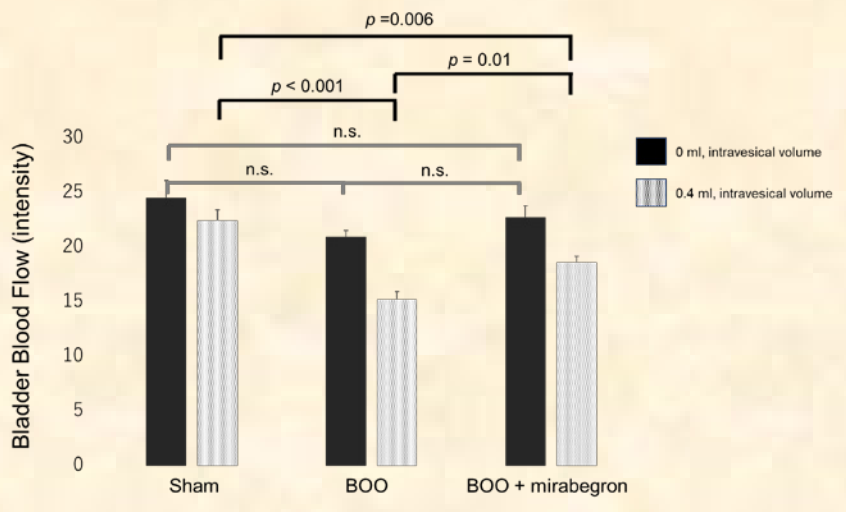
p =0.006
p = 0.01
p < 0.001
n.s.
n.s.
n.s.
0 ml, intravesical volume
0.4 ml, intravesical volume
Bladder Blood Flow (intensity)
30
25
20
15
10
5
0
Sham
BOO
BOO + mirabegron

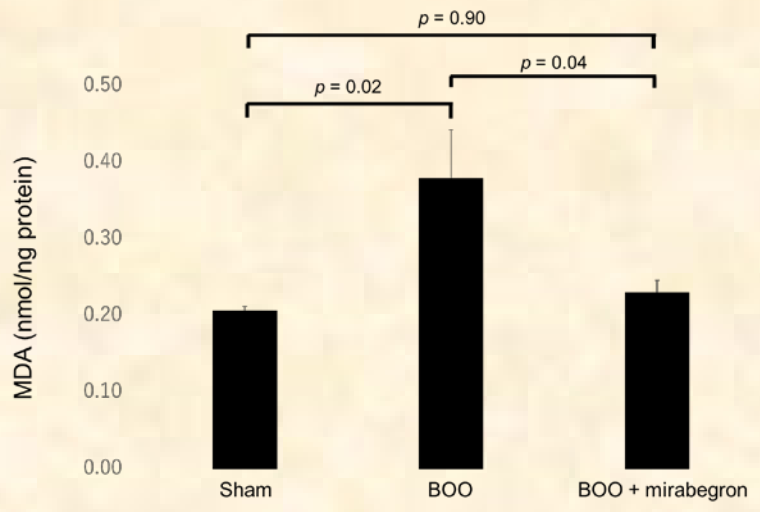
p = 0.90
p = 0.02
p = 0.04
MDA (nmol/ng protein)
0.50
0.40
0.30
0.20
0.10
0.00
Sham
BOO
BOO + mirabegron

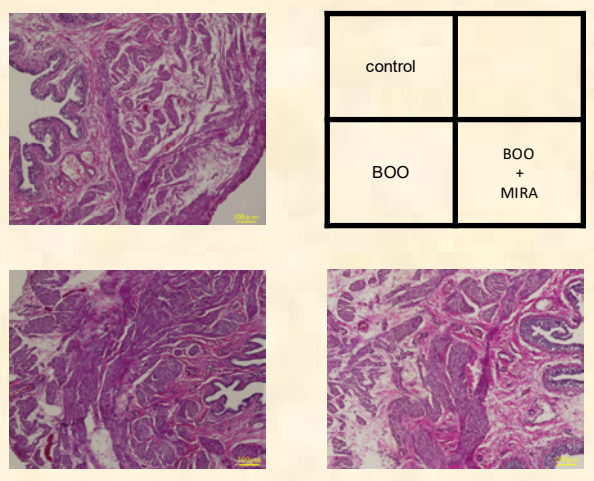
control
BOO
BOO + MIRA

CONCLUSIONS

Mirabegron treatment significantly improved BOO-induced bladder dysfunction through the amelioration of bladder blood flow.

ACKNOWLEDGEMENT

This study was funded by Astellas pharma. Mirabegron was provided by Astellas pharma.